GENE AND EPIGENETIC EXPRESSION PATTERNS OF SAME-GENOME SKIN EQUIVALENT CONSTRUCTS EXPOSED TO SINGLE AND COMBINATIONS OF COSMETIC ACTIVE INGREDIENTS

Bret Stephens[1], Jin Namkoong[2], Dale Kern[2], Remona Gopaul[2], Helen E. Knaggs[2]

[1]Wasatch Scientific Services, Salt Lake City, United States

[2]Center for Anti-Aging Research, Nu Skin Enterprises, Inc., Provo, United States

INTRODUCTION

- *Oryza sativa*, *Narcissus tazetta* bulb and *Schizandra chinensis* fruit extracts are currently used in skin care formulations, however the sub-cellular effects of these extracts on skin cells is poorly understood.
- RNA-seq and ChIP-seq represent contemporary methods to obtain a snapshot of gene expression and epigenetic state.
- Skin equivalent models consisting of adult female matched donor keratinocytes and fibroblasts should more closely replicate skin biology of adult females compared to monolayer culture models or typical commercially constructed tissues containing cells from different donors.

OBJECTIVE

Investigate gene and epigenetic expression patterns of same-genome keratinocyte/fibroblast tissue constructs exposed to *Oryza sativa*, *Narcissus tazetta* bulb and *Schizandra chinensis* fruit extracts individually and in combination.

METHODS

Label	Treatment Conditions
C	Untreated control
A	*Oryza sativa* extract
B	*Narcissus tazetta* bulb extract, *Schizandra chinensis* fruit extract
A+B	Combination of A and B

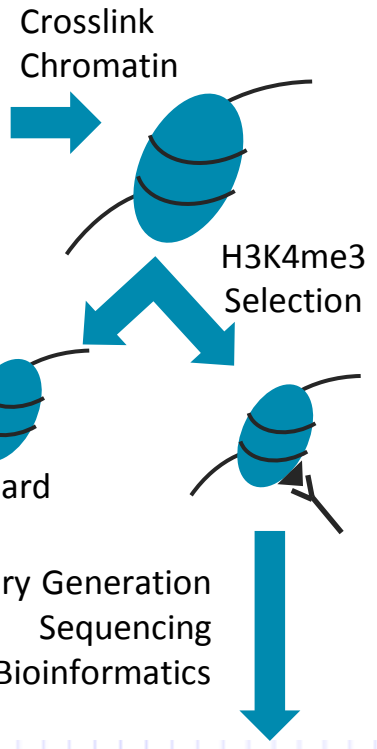
Crosslink
Chromatin
H3K4me3
Selection
Discard
Library Generation
Sequencing
Bioinformatics

RNA Isolation
Library Generation

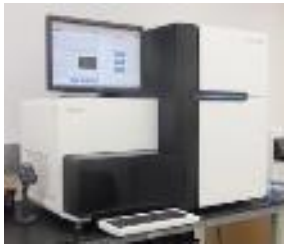

Bioinformatics

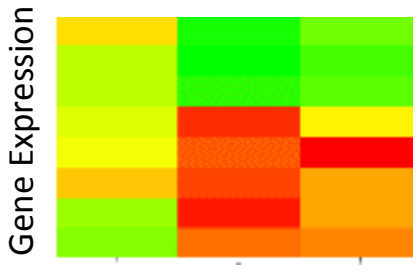
Gene Expression
Treatment Groups

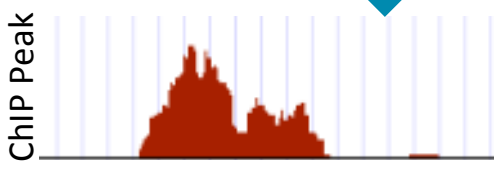
ChIP Peak
Promoter/Genic Region

Compare

RESULTS

I. RNA-seq identifies modulation of genes expression and pathways involved in skin differentiation.

	Genes Expressed	
	≥ 2-fold	≤ 2-fold
Treatment A	81	313
Treatment B	457	1027
Treatment A+B	721	1219

Table A. Overall there were 394, 1484, and 1940 genes differentially regulated at a level of greater than 2-fold induction or repression for treatments A, B, and A+B, respectively. Treatment B had a more dramatic effect on transcriptional changes than treatment A. Interestingly, it was observed that there were significantly more down-regulated genes than up-regulated genes in each treatment. Also of note, the genes differentially expressed in treatment A+B overlapped more closely with treatment B than treatment A, suggesting that the contribution of treatment B is more dominant in the combinatorial treatment.

Top 5 Gene Ontology (GO) Term Biological Processes		
	Treatment A	**Treatment B**
1	Epidermis development	Epidermis development
2	Keratinization	Ectoderm development
3	Ectoderm development	Epidermal cell differentiation
4	Response to wounding	Keratinization
5	Keratinocyte differentiation	Keratinocyte differentiation

Table B. Genes identified as up- or down-regulated were fed into the gene ontology program DAVID[1] (version 6.7). Pathways of highest probability are listed in order. It was noted that treatment B and treatment A+B dramatically reduced the expression of genes located in the epidermal differentiation complex (EDC). The EDC is enriched for genes associated with epidermal terminal differentiation: S100 genes, small proline-rich region (SPRR) genes, and late cornified envelope (LCE) genes[2]. Of note, KLF4 (a master stem cell regulatory transcription factor) controls expression of EDC genes and also was found down-regulated in our samples.

II. ChIP-seq findings enhance expression results.

Peaks defining where DNA was bound to the histone marks of interest (H3K4me3 and H3K9ac) were identified and quality scored. Using the UCSC genome browser it was observed that treatment B and A+B had very similar peak shifts, which differed from the control group in several important genic regions. The KLF4 gene described above is provided as an example for H3K4me3 (top). Importantly, many genes identified through differential analysis with ChIP-seq did not show up in the RNA expression analysis. For example, the epithelial- specific factor EHF scored highly in ChIP-seq (bottom), but not in the RNA-seq. Conversely, it was noted that many differentially regulated genes from the RNA-seq did not have ChIP-seq peaks of significant quality for comparison.

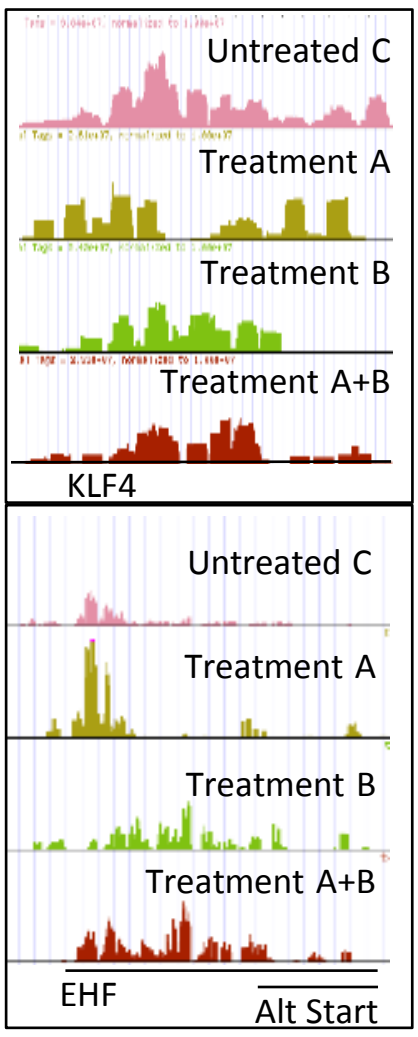
Untreated C
Treatment A
Treatment B
Treatment A+B
KLF4
Untreated C
Treatment A
Treatment B
Treatment A+B
EHF
Alt Start

CONCLUSIONS

To our knowledge, this is the first comprehensive signaling study evaluating the effects of these extracts in a model system intended to best replicate the most common users of cosmetic products by utilizing matched adult female keratinocytes and fibroblasts. The RNA-seq results suggest that these extracts (more particularly *N. tazetta* and *S. chinensis*) reduce expression of many skin-specific factors that play a role in terminal differentiation, leading to an expression profile that mimics youthful skin. As expected, ChIP-seq analysis proved useful in validating RNA-seq findings. More importantly, the ChIP-seq results have exposed additional patterns at potentially key genic regions that were not evident in the RNA-seq data. Conversely, ChIP-seq was not able to identify all differentially regulated transcripts. Therefore, using both platforms in concert can provide additional layers of understanding compared to just a single method. Further mining of this data, combined with follow-up studies are planned.

REFERENCES

[1]Huang et al. Nature Protoc. 2009;4(1):44-57

[2]Henry et al. Front Biosci. 2012;1(17):1517-32

Conflict of Interest

JN, DK, RG and HK are employees of Nu Skin Enterprises, Inc.

BS received funding from Nu Skin Enterprises, Inc.

Contact: Dale Kern, dkern@nuskin.com